Quality of life of treatment for relapsed/refractory multiple myeloma: a systematic review

Maria Nassim[1], Jonas Nilsson[1], Stojan Zavisic[2], Mia Malmenäs[1], Iain Fotheringham[3], Sneha Purushotham[3], Ansa Ibok[3]
[1] ICON plc, [2] Oncopeptides, Stockholm, Sweden, [3] ICON plc, London, United Kingdom

Introduction

- Multiple Myeloma (MM) is an incurable cancer, accounting for 10% of hematologic malignancies with 159,985 newly diagnosed cases in 2018[1] representative of an increasing global incidence. Due to its natural history, most patients eventually relapse and become resistant to therapy.
- The definition of relapsed Refractory Multiple Myeloma (RRMM) varies, across studies, disease information websites and health technology appraisal documents. However, for the purpose of the SLR, the following definition was used: "Patients with RRMM who have had at least one therapy".
- Given the severity of RRMM and its association with severe symptoms impairing Health-Related Quality of Life* (HRQoL), preservation (if not improvement during treatment) becomes an increasingly important aspect.
- HRQoL is widely recognized as an important tool for use in health economic evaluations and is an important measure in assessing the therapeutic value of novel anti-myeloma MM drugs and drug combinations.

Objective

- The objective of this study was to gain understanding of existing peer-reviewed clinical evidence from a therapeutic standpoint for patients with RRMM.
- To attain this objective, a systematic literature review (an SLR) was conducted to evaluate the efficacy and safety of treatments for patients with MM who have experienced relapse or refractory disease after at least one treatment with specific focus on patient HRQoL.

Methods

- An SLR was developed to understand the randomised controlled trial (RCT) evidence base for patients with RRMM then subsequently identify and extract relevant quality of life data.
- The SLR was conducted in accordance with the Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA) statement[4] (see Figure 1).
- The following eligibility criteria were applied, to identify relevant RCT HRQoL data:

Table 1: Eligibility criteria

Inclusion	
Population	Adults (≥18 years) with a diagnosis of RRMM* *"People with relapsed or refractory multiple myeloma (RRMM) who have had at least one therapy
Intervention and Comparators	Pharmacological and non-pharmacological treatments for RRMM
Outcomes	All reported HRQoL, including QLQ-C30, QLQ-MY20 and EQ-5D
Study design	Randomized controlled trials (crossover studies also included) from 2008-current Phase 1 RCTs, pharmaco-economics, observational studies, pharmacokinetics and pharmacodynamics studies, methodology studies or protocols, reviews, and comments/ letters/ were excluded
Date	Full-text articles published 2008 and October 2018 (date of search) Conference proceedings published from 2016 to the October 2018 (date of search)
Language	English language

Results

- In total, 66 articles and 20 conference abstracts reporting results from 47 RCTs were identified in the search (Figure 1), of which only 7 studies reported HRQoL evidence in RRMM patients.

Figure 1: PRISMA flow diagram of SLR

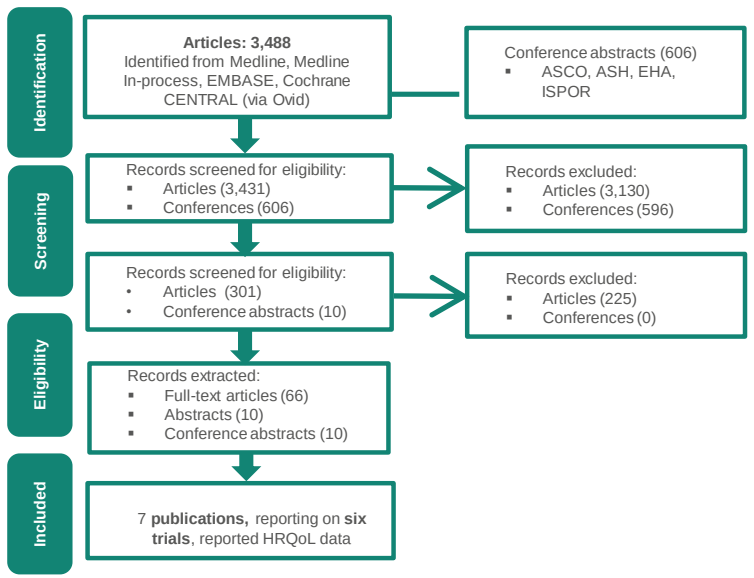
Identification
Screening
Eligibility
Included
Articles: 3,488
Identified from Medline, Medline In-process, EMBASE, Cochrane CENTRAL (via Ovid)
Conference abstracts (606)
ASCO, ASH, EHA, ISPOR
Records screened for eligibility:
Articles (3,431)
Conferences (606)
Records excluded:
Articles (3,130)
Conferences (596)
Records screened for eligibility:
Articles (301)
Conference abstracts (10)
Records excluded:
Articles (225)
Conferences (0)
Records extracted:
Full-text articles (66)
Abstracts (10)
Conference abstracts (10)
7 **publications,** reporting on **six trials**, reported HRQoL data

- Identified RCTs involved an average of 723 patients (with characteristics summarised in Table 2) and covered a total of 7 interventions from 3 different drug classes for RRMM.

Table 2: Key demographic and clinical characteristics across selected studies

Characteristic	Studies reporting characteristic, n	Median	Range
Baseline no. of patients	6	722	455 - 929
Age Median Range	 5 5	 62 -	 61 – 66 33 - 91
Gender (%) Male Female	 5 5	 56.1 44	 54.3 - 59.2 43.8 – 45.7
Male Gender, %	5	56.1	54.3 - 59.2
RRMM Treatment regimen Monotherapy	 1	 1	 --
Combination therapy	6	6	--
Disease duration from diagnosis, median years (range)	1	3.5	0.4 - 24.9
MM Type, % Refractory	 1	 82.4	 -
Relapsed	3	100	--
Refractory or relapsed and refractory	2	51.2	2.4 - 100
Refractory or intolerant	1	15.2	--

Results

- Of the 7 studies selected, none provided the study drug of choice as a first line therapy, with the range of previous treatments varying from 1 - ≥ 2 across trials.
- The most widely studied group of interventions were proteasome inhibitors (PI), monoclonal antibodies (mAbs), and immunomodulatory drugs (IMiDs), represented in 44%, 22% and 15% of the studies, respectively.
- Few studies reported longitudinal HRQoL data. In the few that did, there was inconsistency in HRQoL changes: [3,6,9]. For proteasome inhibitors (PIs) combined with immunomodulatory agents (IMiDs), HRQoL was sustained while maintaining QLQ-C30 GHS/QoL mean scores relatively stable over time.
- In addition, 5 publications used minimal important difference (MID) as a measure of clinically meaningful improvement in HRQoL. In general, improvements were largely unchanged or sustained for PIs meeting the predefined MID (as shown in Figure 2). IMiDs however reported significant clinically meaningful improvements in HRQoL.

Figure 2: Summary of EORTC QLQ-C30 GHS/QoL scores showing changes from baseline values across 7 selected studies

Drug Class & Intervention	Mean EORTC QLQ-C30 GHS/QoL score change from baseline

1) PIs

a) Ixazomib: showed no statistically significant differences compared to control group[10]

b) Carfilzomib: associated with superior HRQoL scores in QLQ-C30 global health status as time progressed[3,8,9]

c) Bortezomib: differences in QLQ-30 global health status scores favored bortezomib over dexamethasone in most of the domains[6].

2) IMiDs

Pomalidomide: elicited clinically meaningful improvements in HRQoL over time with significant enhancements demonstrated in physical functioning, emotional functioning health utility and fatigue scores[5].

3) Pan-HDAC inhibitors

Panobinostat: QLQ-MY20 showed an initial improvement in most domains and subsequent stabilization for both arms[6].

It significantly extended median time to clinically meaningful worsening in HRQoL in almost all functional and symptom domains.

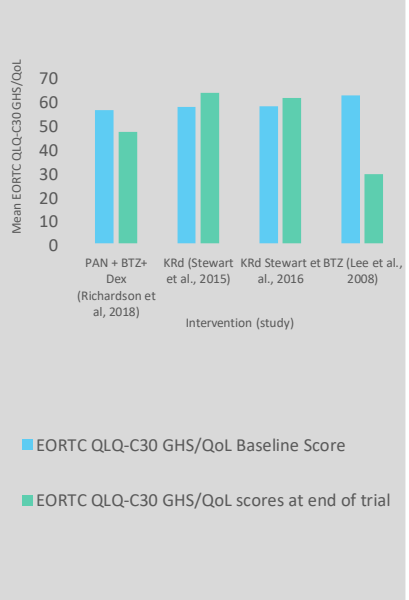
Mean EORTC QLQ-C30 GHS/QoL
70
60
50
40
30
20
10
0
PAN + BTZ+ Dex (Richardson et al, 2018)
KRd (Stewart et al., 2015)
KRd Stewart et al., 2016
BTZ (Lee et al., 2008)
Intervention (study)
EORTC QLQ-C30 GHS/QoL Baseline Score
EORTC QLQ-C30 GHS/QoL scores at end of trial

Discussion

- Reports of substantial impairment to health-related quality of life (HRQoL) including reduced physical function, fatigue, and pain are commonplace in patients with multiple myeloma (MM)
- However, there is a notable lack of published HRQoL data from RCTs.
- Furthermore, existing HRQoL data lacks consistency in HRQoL outcome collection, with only a few of the 7 selected studies (Figure 3) tracking absolute GHS HRQoL scores from baseline (as opposed to merely reporting improvements or differences in HRQoL), thereby limiting comparability across therapies.

- The issue is further confounded by the diversity of patient groups between clinical trials, particularly with regards to pathology and age distribution which may influence HRQoL data.
- While there will no doubt be many more non-RCT studies available with HRQoL data, it is important to initially understand the impact that therapies have on patient HRQOL in a controlled setting
- Tools utilised to evaluate quality of life and corresponding methodology varied across the 7 selected studies and only a small number provided detailed EORTC-QLQ-C30, EORTC-QLQ-MY20, FACT/GOG-Ntx and EQ-5D results.
- QLQ-C30, which attempts to assess multiple facets of patient health, was reported in all seven studies.
- The QLQ-C30 data suggests that some therapies may at least sustain patient HRQoL within the timeframe of the RCTs.

Figure 3: Reported EORTC QLQ-C30 GHS HRQoL scores measures over time

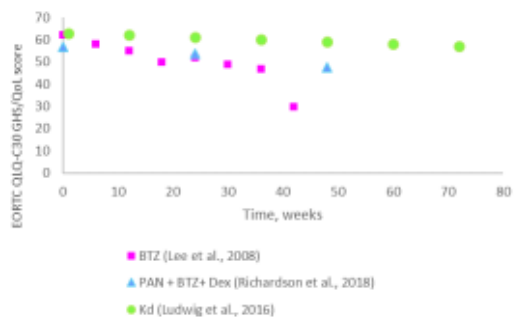
EORTC QLQ-C30 GHS/QoL score
70
60
50
40
30
20
10
0
0
10
20
30
40
50
60
70
80
Time, weeks
BTZ (Lee et al., 2008)
PAN + BTZ+ Dex (Richardson et al., 2018)
Kd (Ludwig et al., 2016)

Conclusion

- Despite a wealth of RCT evidence assessing the efficacy and safety of therapies for patients with RRMM, there is a notable paucity of HRQoL data
- This affects our ability to understand the complete impact therapies may have on patients
- From the available data, there may well be differences in how therapies affect patient HRQoL. However, in the absence of thorough, comparable information. this is difficult to confidently ascertain. Therefore, the necessity for generation of further data, to enable comparison across therapies, is vital for greater understanding of the true impact on patient quality of life.

References
1. Bray, F. , Ferlay, J. , Soerjomataram, I. , Siegel, R. L., Torre, L. A. and Jemal, A. (2018), Global cancer statistics 2018: GLOBOCAN estimates of incidence and mortality worldwide for 36 cancers in 185 countries. CA: A Cancer Journal for Clinicians, 68: 394-424.
2. Nooka, A. K., Kastritis, E., Dimopoulos, M. A., & Lonial, S. (2015). Treatment options for relapsed and refractory multiple myeloma. Blood, 125(20), 3085-3099.
3. Stewart AK, Rajkumar SV, Dimopoulos MA, Masszi T, Spicka I, Oriol A, et al. Carfilzomib, lenalidomide, and dexamethasone for relapsed multiple myeloma. New england journal of medicine. 2015;372(2):142-52.
4 Liberati A, Altman DG, Tetzlaff J, Mulrow C, Gotzsche PC, Ioannidis JP, et al. The PRISMA statement for reporting systematic reviews and meta-analyses of studies that evaluate health care interventions: explanation and elaboration. J Clin Epidemiol. 2009;62(10):e1-34.
5. Weisel K, Dimopoulos M, Song KW, Moreau P, Palumbo A, Belch A, et al. Pomalidomide and Low-Dose Dexamethasone Improves Health-Related Quality of Life and Prolongs Time to Worsening in Relapsed/Refractory Patients With Multiple Myeloma Enrolled in the MM-003 Randomized Phase III Trial. Clinical lymphoma, myeloma & leukaemia. 2015;15(9 // (GSK) *Novartis* // *Novartis*):519-30
6. Richardson PG, Schlossman RL, Roy AN, Panneerselvam A, Acharyya S, Sopala M, et al. Patient-reported outcomes of multiple myeloma patients treated with panobinostat after >=2 lines of therapy based on the international phase 3, randomized, double-blind, placebo-controlled PANORAMA-1 trial. British journal of haematology. 2018;181(5):628-36.
7. Lee SJ, Richardson PG, Sonneveld P, Schuster MW, Irwin D, San Miguel JF, et al. Bortezomib is associated with better health-related quality of life than high-dose dexamethasone in patients with relapsed multiple myeloma: results from the APEX study. British journal of haematology. 2008;143(4):511-9.
8. Ludwig H, Moreau P, Dimopoulos MA, Mateos MV, Kaiser MF, Feng S, et al. Health related quality of life results from the open-label, randomized, phase III endeavor trial evaluating carfilzomib and dexamethasone versus bortezomib and dexamethasone in patients with relapsed or refractory multiple myeloma. Blood Conference: 58th annual meeting of the american society of hematology, ASH. 2016;128(22)
9. Stewart AK, Dimopoulos MA, Masszi T, Spicka I, Oriol A, Hajek R, et al. Health-related quality-of-life results from the open-label, randomized, phase III ASPIRE trial evaluating carfilzomib, lenalidomide, and dexamethasone versus lenalidomide and dexamethasone in patientswith relapsed multiple myeloma. Journal of clinical oncology. 2016;34(32):3921-30
10. Leleu X, Masszi T, Bahlis NJ, Viterbo L, Baker B, Gimsing P, et al. Patient-reported health-related quality of life from the phase III TOURMALINE-MM1 study of ixazomib-lenalidomide-dexamethasone versus placebo-lenalidomide-dexamethasone in relapsed/refractory multiple myeloma. American Journal of Hematology. 2018;04:04.